Retinal capillary changes and cytokine expression in Spontaneously Diabetic Torii fatty rats

Kasumi Kikuchi[1)], Miyuki Murata[1)], Yasushi Kageyama[2)], Masami Shinohara[2)], Tomohiko Sasase[3)], Kousuke Noda[1)], Susumu Ishida[1)]

[1)] Department of Ophthalmology, Faculty of Medicine and Graduate School of Medicine, Hokkaido University, Sapporo, Japan

[2)] Tokyo Animal & Diet Department, CLEA Japan, Inc., Tokyo, Japan

[3)] Biological/Pharmacological Research Laboratories, Central Pharmaceutical Research Institute, Japan Tobacco Inc., Osaka, Japan

Presentation Number-Poster Board Number
#279-F0324

Background

Spontaneously Diabetic Torii (SDT) rats

- SDT rat is an inbred strain of Sprague Dawley (SD) rat and was established as a non-obese model of type II diabetes.[1]
- SDT rat develops diabetic cataract at 40 weeks of age and diabetic retinopathy (DR) at 70 weeks of age. [2]

SDT fatty rats

- SDT fatty rat was established by introducing the *fa allele* of the leptin receptor gene of Zucker fatty rat into SDT rat genome.
- SDT fatty rat exhibits overt obesity, hyperglycemia and hyperlipidemia at a young age because of polyphagia.
- With an early onset of diabetes mellitus, diabetic complications were observed at a young age.[3]
- SDT fatty rat develops cortical cataract and posterior capsular opacification, commonly observed in patients with type II diabetes, from 7 weeks of age.[4]

→ However, retinal changes in SDT fatty rats have not yet been fully investigated.

Purpose

To investigate the diabetes-induced pathological changes in the retina of SDT fatty rats

Materials & Methods

Animals

Male SDT fatty rats and age-matched SD rats were used in this study.

Capillary diameter

The trypsin-digested retinal vessels were stained with hematoxylin and eosin (H&E). Capillary diameter was measured at a distance of 1500 µm from the optic nerve head.

Pericyte/endothelial cell (P/E) ratio

Retinal capillary network isolated by trypsin digestion was immunostained using anti-NG2 antibody. The total number of cells of the blood vessels was counted and divided into NG2-positive cells (pericytes) and NG2-negative cells (endothelial cells). The P/E ratio was calculated.

Immunostaining for leukocyte adhesion molecules

The trypsin-digested specimens were immunostained using the following antibodies.

- Mouse monoclonal anti-ICAM1 (1:100, 1A29, Novus biological, Centennial, CO, USA)
- Rabbit monoclonal anti-VCAM1 (1:50, EPR5047, Abcam, Cambridge, UK)

DNA microarray

Total RNA was collected from the retinas of SD rats and SDT fatty rats using NucleoSpin RNA Plus and microarray analysis was performed.

Quantitative real-time PCR

The mRNA expression levels of *Tnf*, *Il1b*, *Ccl2/Mcp1*, *Icam1*, *Vegf* and *Il6* were examined by quantitative real-time PCR.

Magnetic Luminex assay

The protein levels of TNF-α, IL-1β, MCP-1, ICAM-1, VEGF-A and IL-6 in the retinas were measured using magnetic Luminex assay. All results were standardized to the protein concentration measured by BCA protein assay kit.

ICAM-1, intercellular adhesion molecule-1; VCAM-1, vascular cell adhesion molecule-1
MCP-, monocyte chemoattractant protein-1; IL, interleukin; TNF-α, tumor necrosis factor-α
VEGF-A, vascular endothelial growth factor-A

Results

Capillary diameter

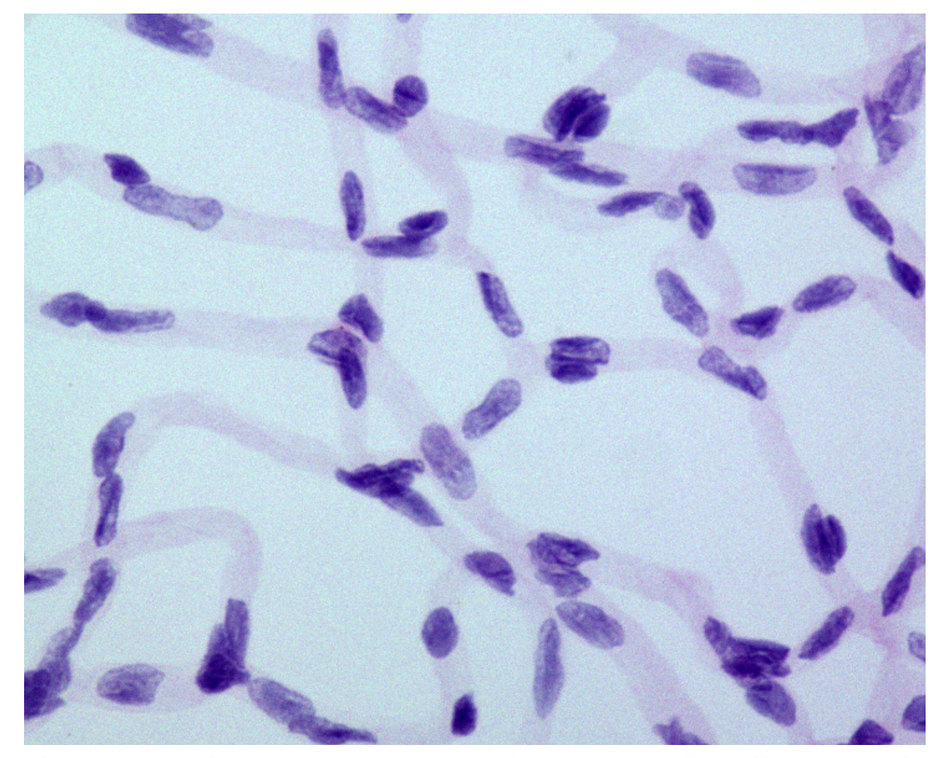

Image of a representative trypsin-digested retinal capillaries (24w SDT fatty rat)

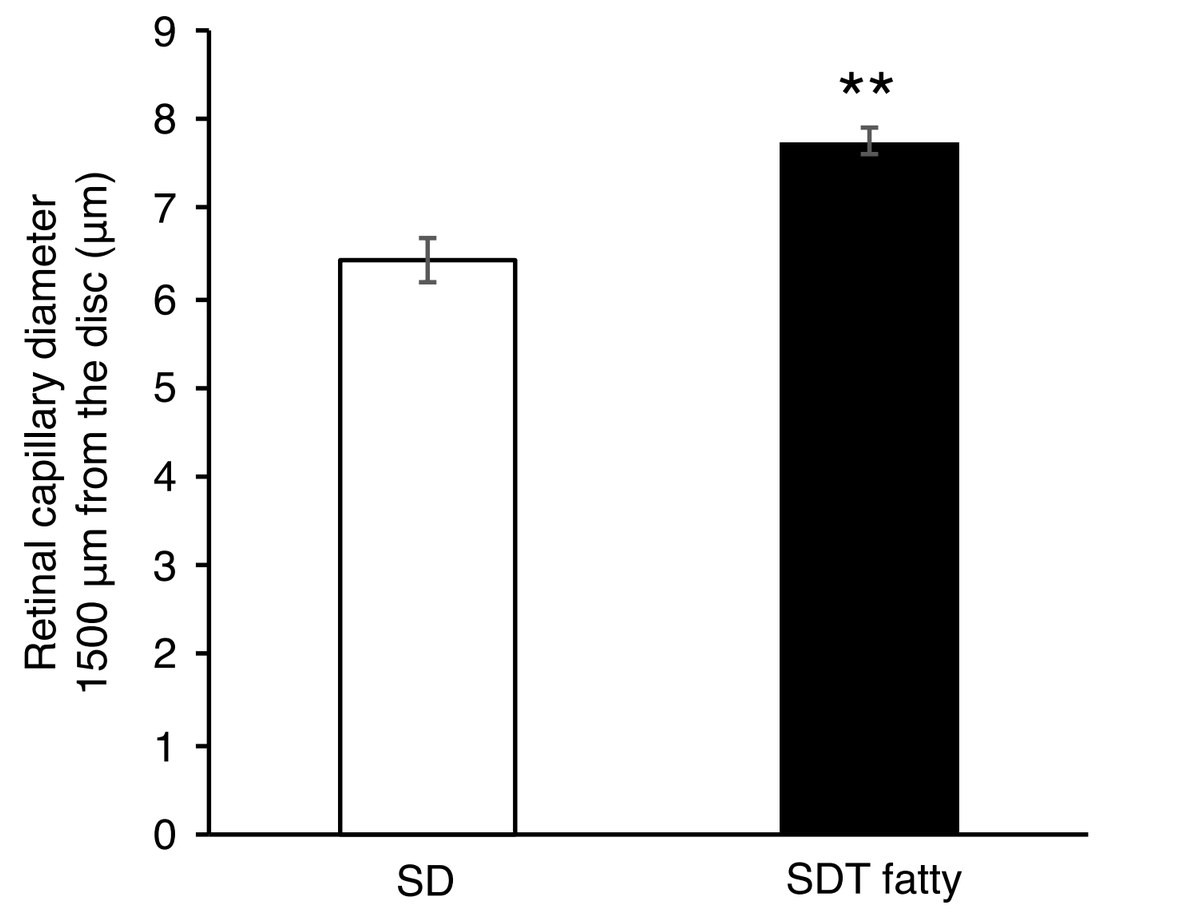

Retinal capillary diameter 1500 µm from the disc (µm)
9
8
7
6
5
4
3
2
1
0
**
SD
SDT fatty

(n=5 each, **P<0.01)

Capillary diameter significantly increased in SDT fatty rats compared to SD rats at 24 weeks of age.

P/E ratio

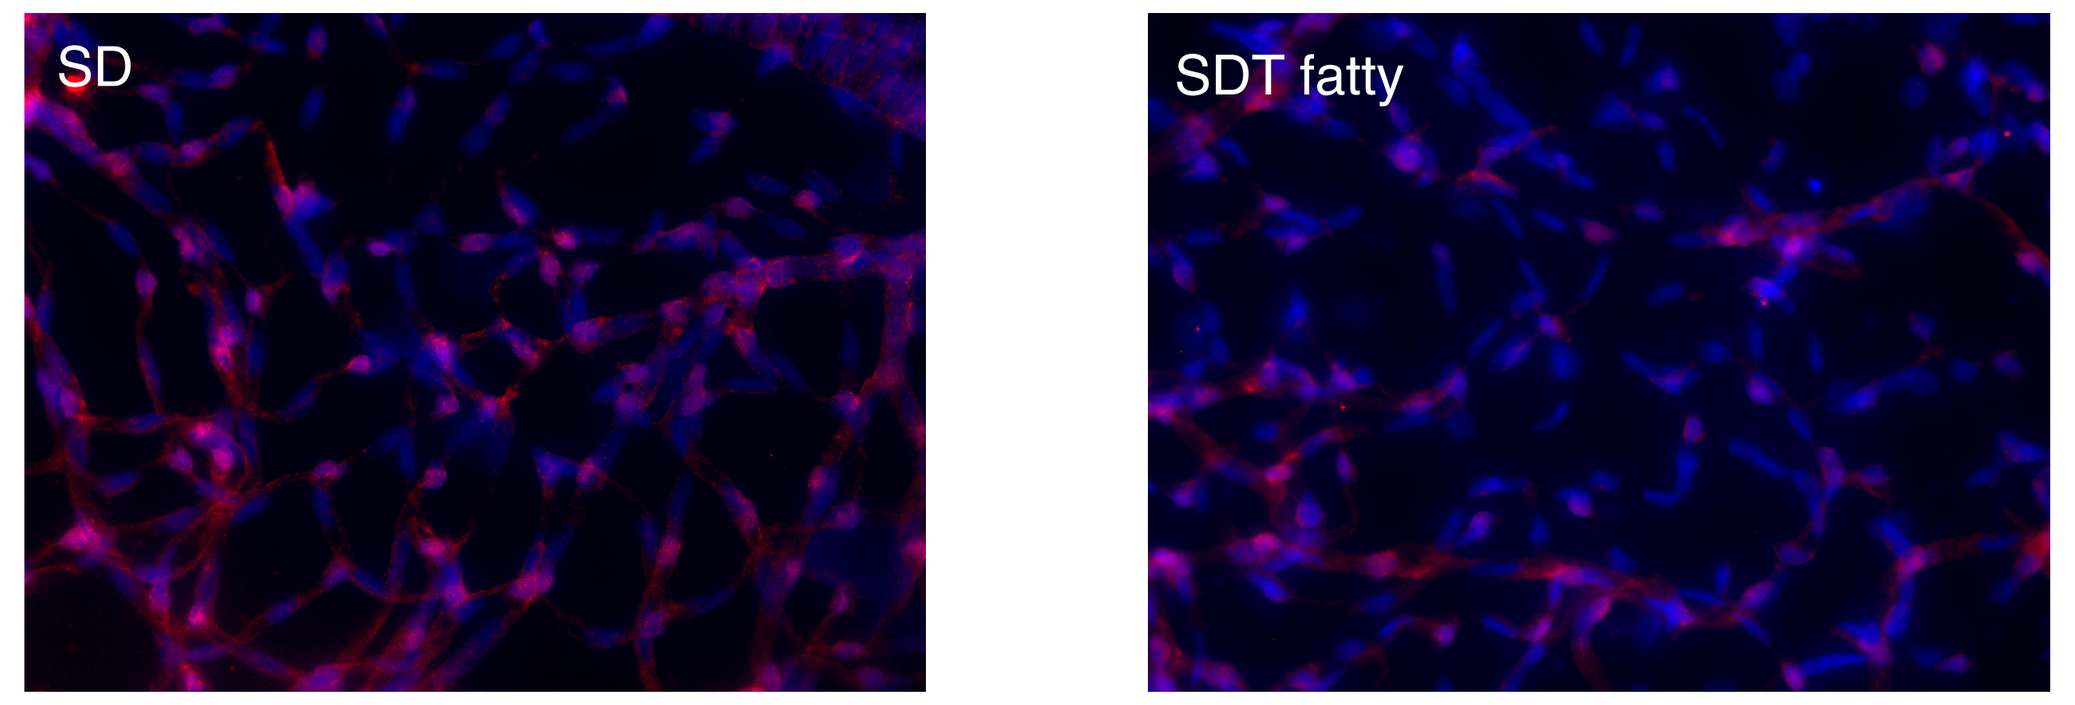

SD
SDT fatty

Red : NG-2 (pericyte marker)
Blue : DAPI (nuclear staining)

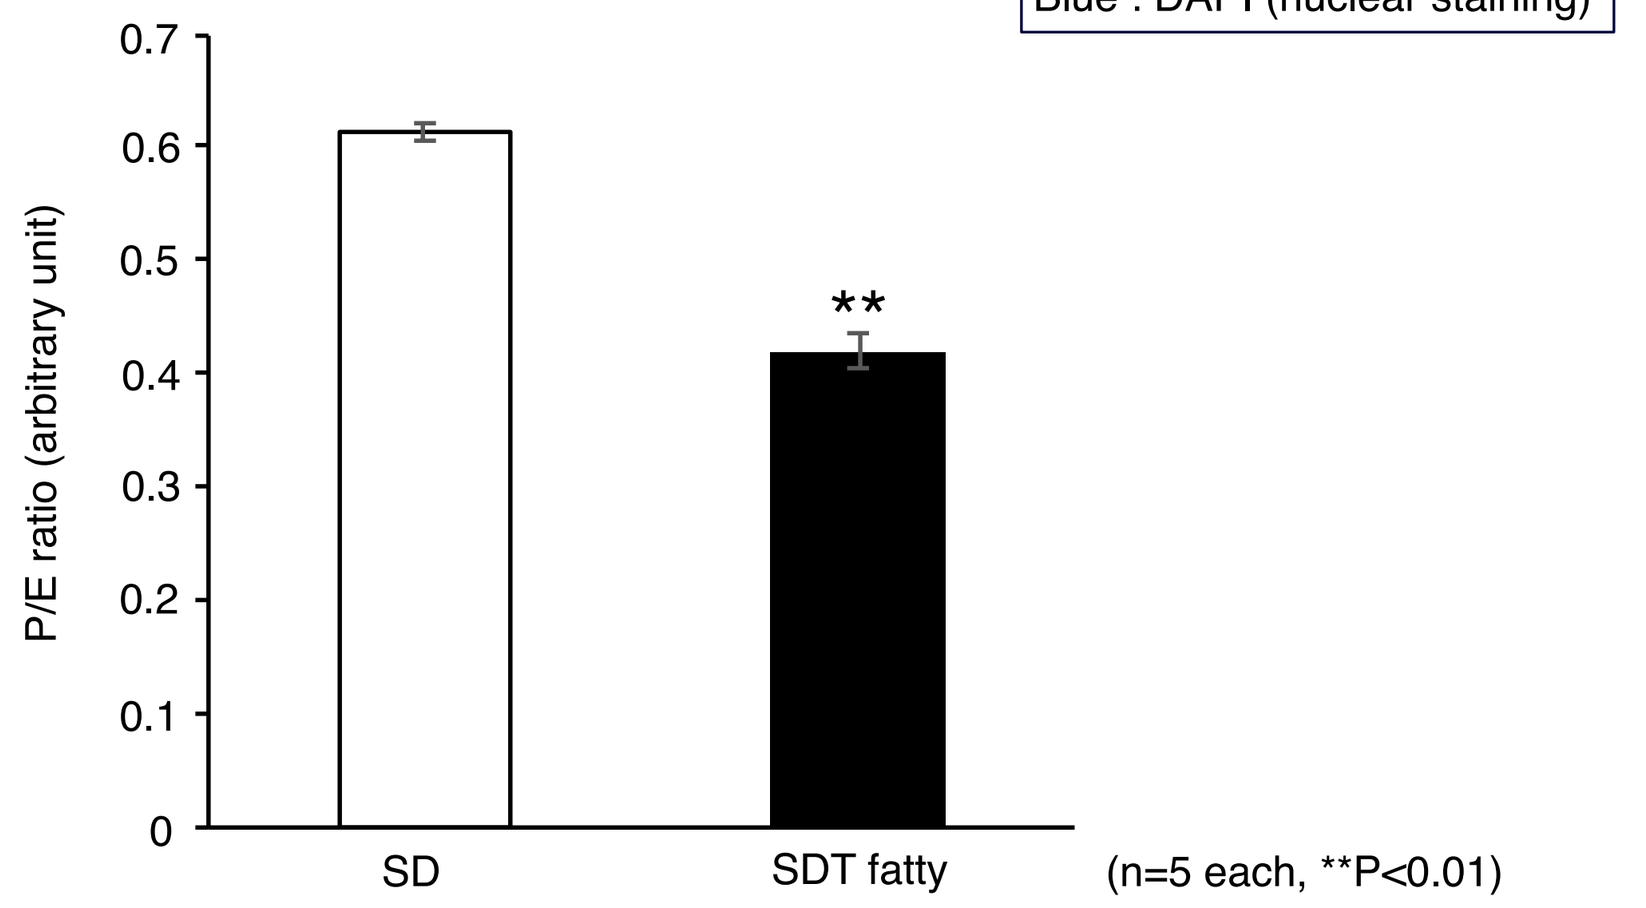

P/E ratio (arbitrary unit)
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0
**
SD
SDT fatty

(n=5 each, **P<0.01)

P/E ratio significantly decreased in the retinal capillaries of SDT fatty rats compared with the age-adjusted controls at 24 weeks of age.

Immunostaining for leukocyte adhesion molecules

SD rat at 24 weeks of age

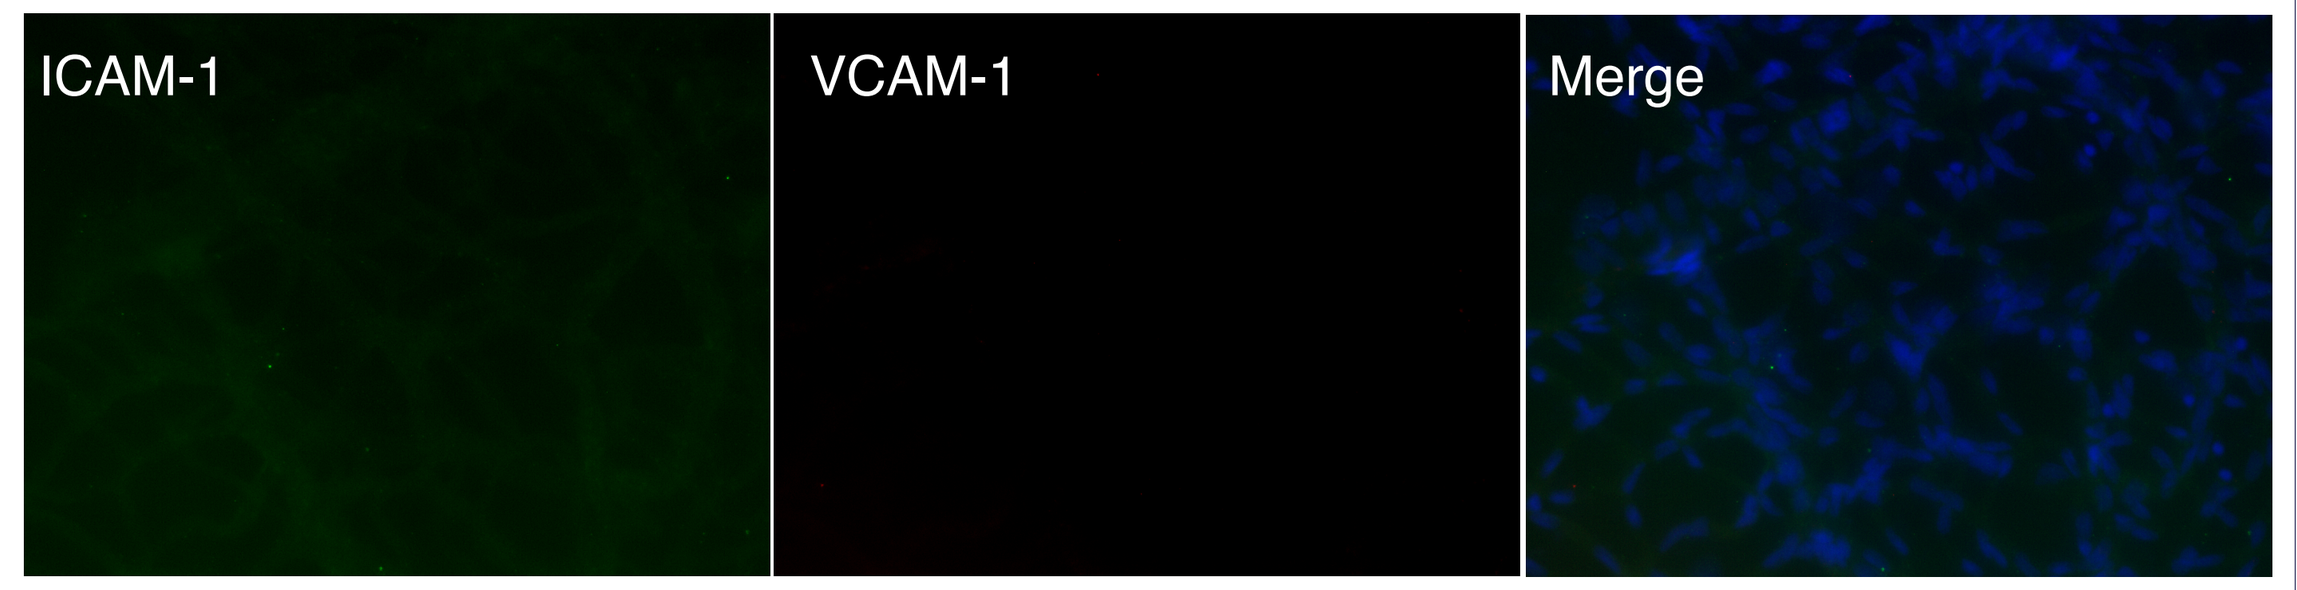

ICAM-1
VCAM-1
Merge

SDT fatty rat at 24 weeks of age

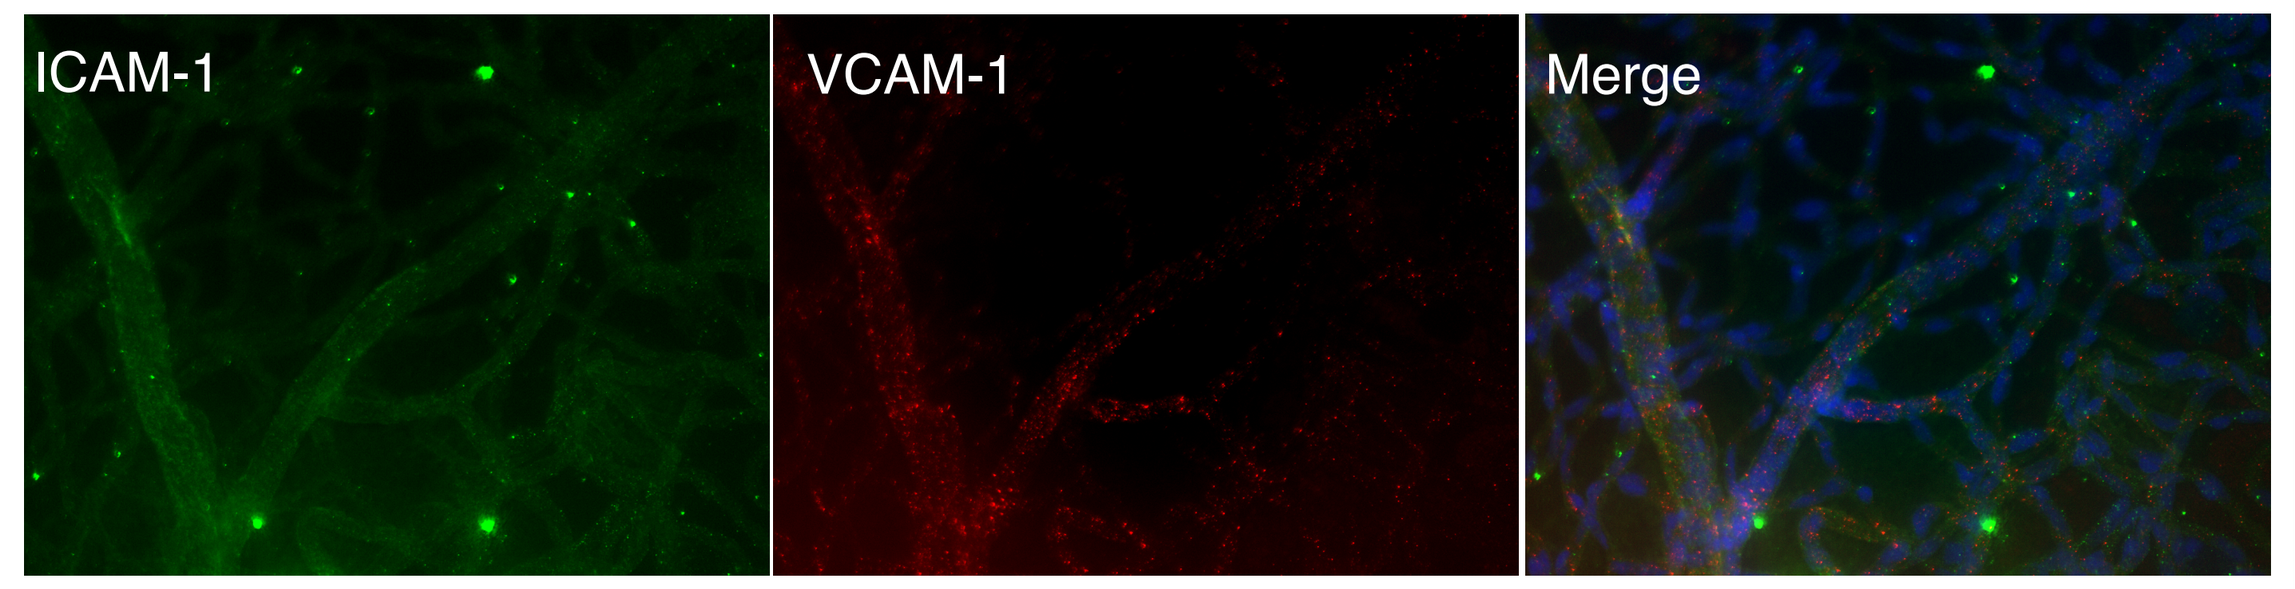

ICAM-1
VCAM-1
Merge

Staining pattern of ICAM-1 and VCAM-1 was faint in SD rats. In contrast, both ICAM-1 and VCAM-1 were explicitly stained along the retinal vessels of SDT fatty rats at 24 weeks of age.

Results

DNA microarray

GeneSymbol	SD signal	SDT fatty signal	Ratio	GenbankAccession
Tnf	24.3	60.1	2.47	NM_012675
Il1b	16.6	56.5	3.40	NM_031512
Mcp1	4.1	66.3	16.13	NM_031530
Icam1	293.0	952.5	3.25	NM_012967
Vegf	10602.8	11077.9	1.04	NM_001287107
Il6	2.9	2.3	0.80	NM_012589

Tnf, Il1b, *Mcp1* and *Icam1* were upregulated more than 2-fold in the retina of SDT fatty rats compared to those of SD rats at 24 weeks of age. By contrast, the expression levels of *Vegf* and *Il6* were not upregulated in the retinas of SDT fatty rats.

Quantitative real-time PCR

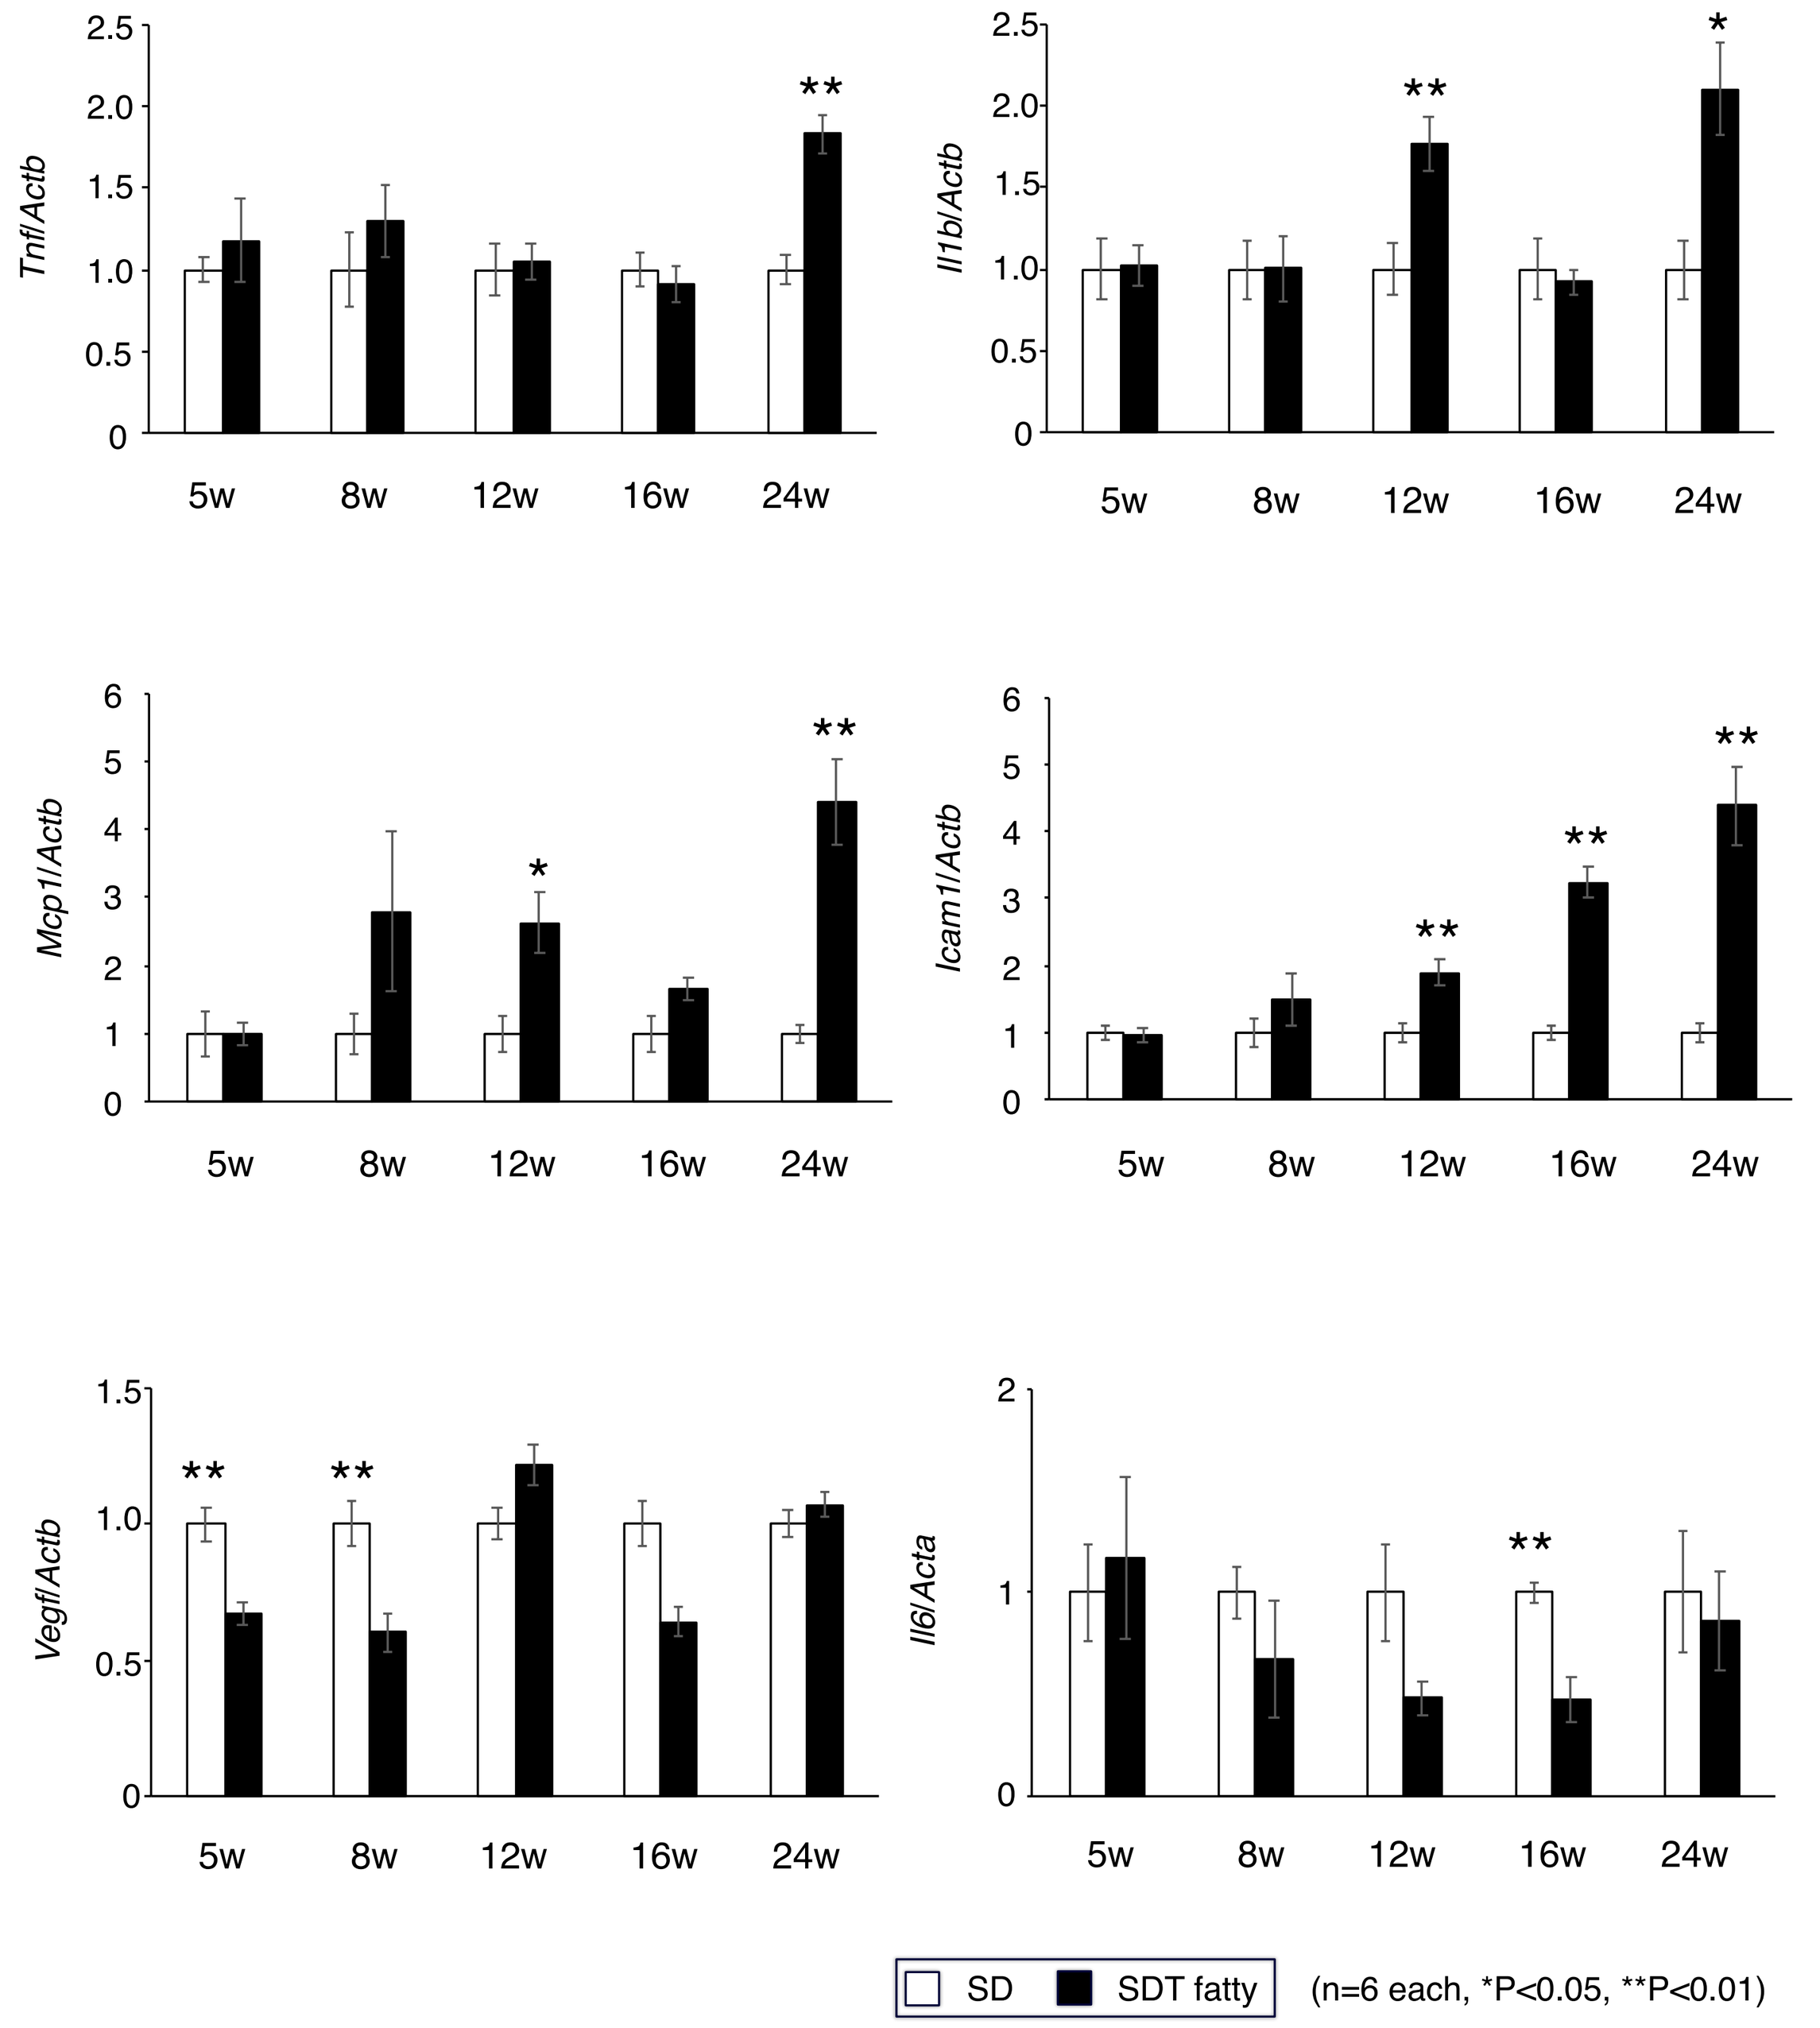

Tnf/Actb
Il1b/Actb
Mcp1/Actb
Icam1/Actb
Vegf/Actb
Il6/Acta
5w
8w
12w
16w
24w
SD
SDT fatty

(n=6 each, *P<0.05, **P<0.01)

Consistent with the results of microarray analysis, the mRNA expression levels of *Tnf, Il1b* and *Mcp1* were significantly upregulated in the retina of 24-week-old SDT fatty rats. *Icam1* mRNA expression showed the gradual increase with age. The mRNA expression levels of *Vegf* and *Il6* were not upregurated, which was also consistent with the results of microarray analysis.

Magnetic Luminex assay at 24 weeks of age

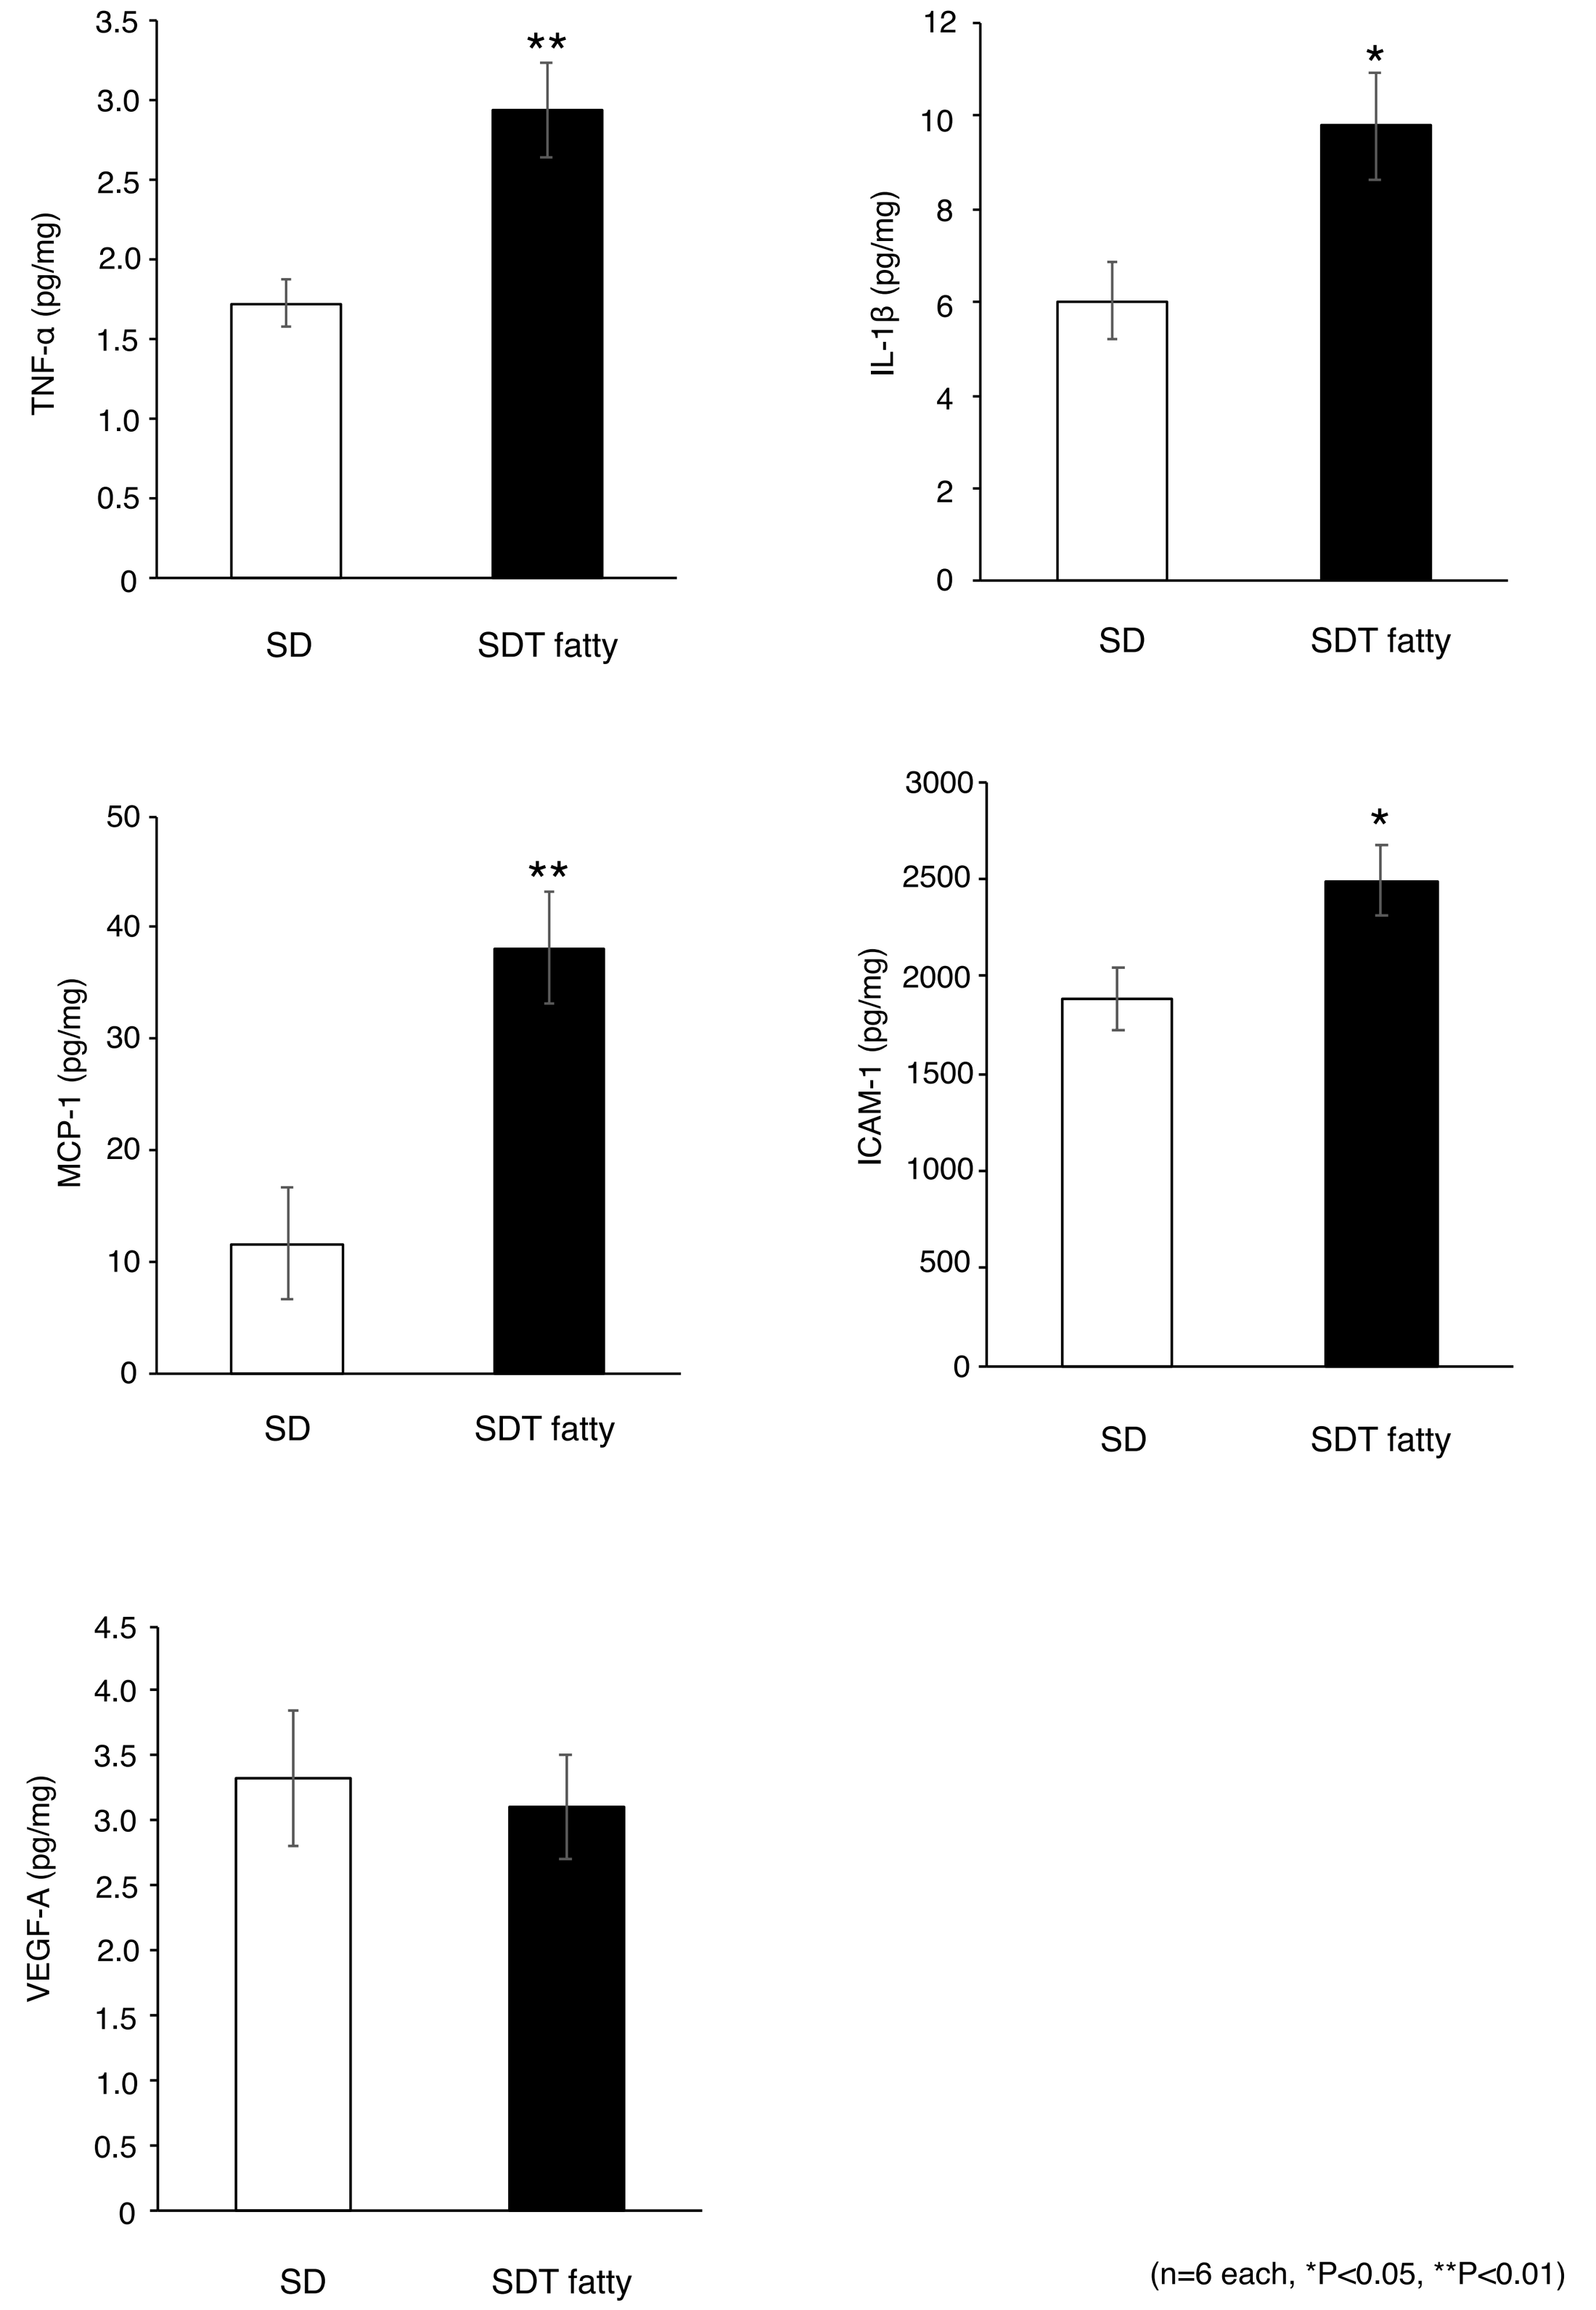

TNF-α (pg/mg)
IL-1β (pg/mg)
MCP-1 (pg/mg)
ICAM-1 (pg/mg)
VEGF-A (pg/mg)
SD
SDT fatty

(n=6 each, *P<0.05, **P<0.01)

TNF-α, IL-1β, MCP-1 and ICAM-1 protein levels were also significantly increased in SDT fatty rats compared to those of SD rats. By contrast, VEGF-A protein level was not significantly different between SD and SDT fatty rats. IL-6 protein level was below the detection limit of the assay in both SD and SDT fatty rats (data not shown).

Summary

- Retinal capillaries of 24-week-old SDT fatty rats were dilated.
- The P/E ratio was reduced in retinal capillaries of 24-week-old SDT fatty rats, indicating the accelerated pericyte loss, an early change of DR.
- Immunostaining revealed that ICAM-1 and VCAM-1 were induced in the retinal capillaries of 24-week-old SDT fatty rats.
- In the retina of 24-week-old SDT fatty rat, both mRNA and protein levels of inflammation-associated molecules, *i.e.*, TNF-α, IL-1β, MCP-1 and ICAM-1, were increased.
- In contrast, VEGF-A and IL-6 were not induced in the retina of 24-week-old SDT fatty rat.

Discussion

Microvascular alterations in SDT fatty rats

Capillary alteration is a hallmark feature of DR. Pericyte loss is a well-known early change in human DR.[5] An et al. examined retinas from patients with non-proliferative DR using a confocal scanning laser microscope and revealed capillary dilation.[6] SDT fatty rats showed capillary alteration similar to these early DR changes in humans.

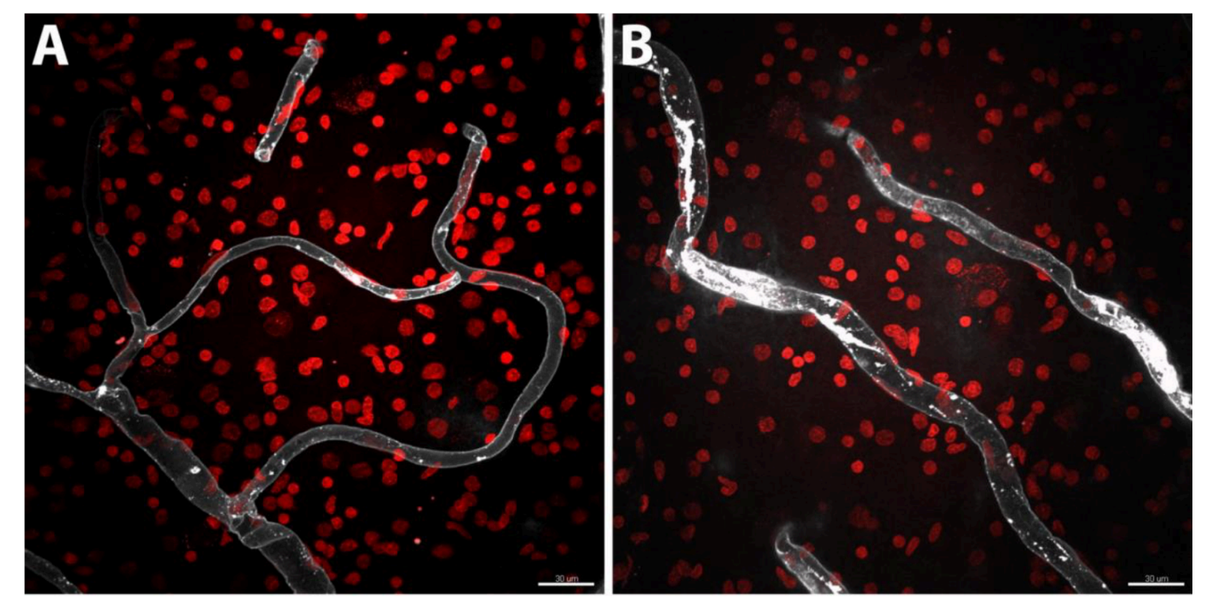

A
B

control DR

(An D et al. *Transl Vis Sci Technol.* 2021)

Leukocyte adhesion molecules in SDT fatty rats

Previous studies have indicated that leukocytes play an important role in the development of DR.[7] Among leukocyte adhesion molecules, we focused on ICAM-1 and VCAM-1 in the present study. Both of ICAM-1 and VCAM-1, the molecules modulating leukocyte adhesion during leukocyte recruitment cascade, have been detected in epiretinal membranes in patient with proliferative DR (PDR),[8] and ICAM-1 has been reported to be induced in the retinal vessels of patients with DR.[9] The present study revealed that ICAM-1 and VCAM-1 were induced along with retinal vasculature in SDT fatty rats.

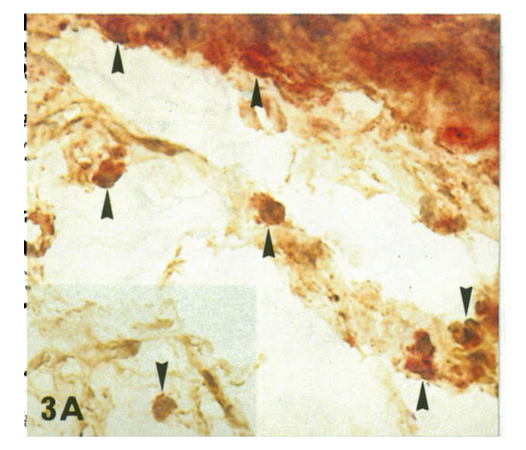

3A

Expression of ICAM-1 in PDR epiretinal membranes[8]

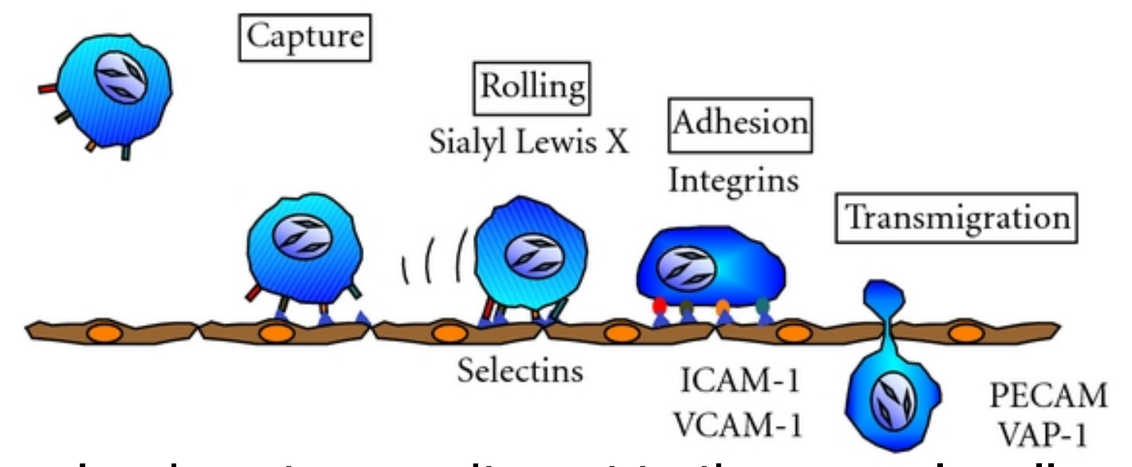

Capture
Rolling
Sialyl Lewis X
Adhesion
Integrins
Transmigration
Selectins
ICAM-1
VCAM-1
PECAM
VAP-1

Leukocyte recruitment to the vessel wall

(Noda K et al. *J Ophthalmol.* 2012)

Inflammatory response in the retinas of SDT fatty rats

DR is clinically divided into two stages based on the proliferative status of the retinal vasculature, that is, the non-proliferative stage and the proliferative stage. Recent studies have elucidated that chronic inflammation underlies much of the vascular complications of DR.[10,11] In the retina of 24-week-old SDT fatty rat, the levels of inflammation-related molecules including TNF-α, IL-1β, MCP-1 and ICAM-1 were elevated, while VEGF-A, which is elevated under ischemic conditions, was not.

Considering that capillary dilation and pericyte loss were the representative morphological findings in the retina of SDT fatty rats, it is plausible that SDT fatty rat is an animal model of early stage of DR.

Conclusions

The present study demonstrated that SDT fatty rats exhibited early diabetic changes in the retina, indicating that SDT fatty rats serve as a potential animal model in researches on the pathogenesis of early human DR.

References

1. Sasase T et al. *J Diabetes Res.* 2013
2. Shinohara M et al. *Int J Exp Diabetes Res.* 2000
3. Matsui K et al. *Exp Anim.* 2008
4. Kikuchi K et al. *J Diabetes Res.* 2020
5. Cogan DG et al. *Arch Ophthalmol.* 1961
6. An D et al. *Transl Vis Sci Technol.* 2021
7. Chibber R et al. *Curr Diabetes Rev.* 2007
8. Tang S et al. *Br J Ophthalmol.* 1994
9. McLeod DS et al. *Am J Pathol.* 1995
10. Adamis AP. *Br J Ophthalmol.* 2002
11. Capitão M et al. *J Cell Biochem.* 2011

Conflict of Interest

Yasushi Kageyama : 【E】CLEA Japan Inc.
Masami Shinohara : 【E】CLEA Japan Inc.
Tomohiko Sasase : 【E】Japan Tobacco Inc.